Abstract #TPS1107

PRESERVE 2: A PHASE 3, RANDOMIZED, DOUBLE-BLIND TRIAL OF TRILACICLIB VERSUS PLACEBO IN PATIENTS RECEIVING FIRST- OR SECOND-LINE GEMCITABINE AND CARBOPLATIN FOR LOCALLY ADVANCED UNRESECTABLE OR METASTATIC TRIPLE-NEGATIVE BREAST CANCER

Shom Goel[1]; Joyce O'Shaughnessy[2]; Antoinette R. Tan[3]; Boris Krastev[4]; Hope Rugo[5]; Philippe Aftimos[6]; Denise A. Yardley[7]; Zoran Andric[8]; Curt Wolfgang[9]; Jessica A. Sorrentino[9]; Wenli Tao[9]; Andrew Beelen[9]; Rajesh Malik[9]; and Sarika Jain[9]

[1] Peter MacCallum Cancer Centre, Melbourne, Australia; [2] Baylor University Medical Center, Texas Oncology, US Oncology, Dallas, TX, USA; [3] Levine Cancer Institute, Atrium Health, Charlotte, NC, USA; [4] MHAT Hospital for Women Health Nadezhda, Sofia, Bulgaria; [5] University of California San Francisco Comprehensive Cancer Center, San Francisco, CA, USA; [6] Institut Jules Bordet, Université Libre de Bruxelles, Brussels, Belgium; [7] Sarah Cannon Research Institute and Tennessee Oncology, Nashville, TN, USA; [8] Clinical Hospital Centre Bezanijska Kosa, Belgrade, Serbia; [9] G1 Therapeutics, Research Triangle Park, NC, USA

INTRODUCTION

- Chemotherapy, alone or in combination with immune checkpoint inhibitors, is the standard of care for patients with metastatic triple-negative breast cancer (mTNBC)[1–3]
 - However, not all patients with programmed death-ligand 1 (PD-L1)–positive TNBC are appropriate candidates for immune checkpoint inhibitor treatment, and some patients with PD-L1–negative TNBC may not derive clinical benefit[4]
 - Additionally, chemotherapy-induced immunosuppression may affect antitumor efficacy, owing to an inability of the host immune system to effectively mount a response against the cancer[5]

Trilaciclib Mechanism of Action

Trilaciclib (IV myeloprotection therapy)

- IV inhibitor of CDK4/6, administered prior to chemotherapy[6]
- Transiently arrests HSPCs and immune cells in the G1 phase of the cell cycle during chemotherapy exposure, protecting them from chemotherapy-induced damage[6–11]

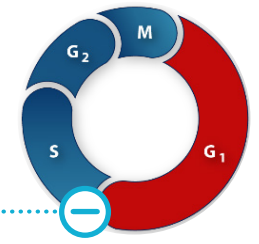

M
G2
S
G1

Myeloprotection impact[6–9]		Antitumor efficacy impact[12–14]
Reduces rate of hematologic adverse events (less neutropenia, anemia, and thrombocytopenia)	Protects multiple myeloid cell lineages	**Increases** patients' ability to receive longer duration of chemotherapy-based regimens
Decreases rescue interventions (fewer transfusions, less G-CSF, and fewer hospitalizations)	Protects lymphoid cell lineages	**Protects** the immune system from damage by chemotherapy
Improves patients' quality of life (improved well-being and less fatigue)	Improves immune response	**Enhances** T-cell activation and favorably alters the tumor microenvironment

CDK4/6, cyclin-dependent kinase 4/6; G-CSF, granulocyte colony-stimulating factor; HSPC, hematopoietic stem and progenitor cell; IV, intravenous.

- Trilaciclib is an intravenous (IV) cyclin-dependent kinase (CDK)4/6 inhibitor indicated to decrease the incidence of chemotherapy-induced myelosuppression in adult patients when administered prior to a platinum/etoposide- or topotecan-containing chemotherapy regimen for extensive-stage small cell lung cancer[6]
- A randomized, open-label, phase 2 trial (NCT02978716) compared trilaciclib prior to gemcitabine plus carboplatin (GCb) with GCb alone in patients with TNBC[13]
 - Although the primary endpoint of myeloprotection was not met, a clinically meaningful improvement in overall survival (OS) was observed in the intention-to-treat population with both PD-L1–positive and –negative tumors[13,14]
 - Among all patients, median OS was 20.1 months with trilaciclib plus GCb versus 12.6 months with GCb alone (hazard ratio 0.36) in the primary analysis[13]

PRESERVE 2 STUDY

- PRESERVE 2 (NCT04799249) is a phase 3, multicenter, randomized, double-blind, placebo-controlled, 2-cohort study evaluating the safety and efficacy of trilaciclib versus placebo administered prior to GCb for patients receiving first- or second-line treatment for advanced/metastatic TNBC
- This study is designed to confirm the OS benefit seen in the phase 2 study, and to evaluate efficacy in a post–checkpoint inhibitor population

STUDY OBJECTIVES

1 Primary Objective
To evaluate the effect of trilaciclib prior to GCb versus placebo prior to GCb on OS

2 Key Secondary Objective
To assess the effect of trilaciclib versus placebo on patients' quality of life, as measured by time to first confirmed deterioration of fatigue (TTCD-fatigue)

STUDY DESIGN

Patients

Cohort 1 (n = 170)[a]
Patients receiving first-line GCb
(PD-1/PD-L1 inhibitor–naïve population)

Stratification factors:
- PD-L1 status
- Disease-free interval
- Country

Cohort 2 (n = 80)
Patients receiving second-line GCb
(previously treated with a PD-1/PD-L1 inhibitor)

No stratification factors

Randomization 1:1

Treatment phase

Trilaciclib prior to GCb on days 1 and 8 every 21 days

Placebo prior to GCb on days 1 and 8 every 21 days

Continue until:
- Unacceptable toxicity
- Progressive disease
- Withdrawal of consent
- Discontinuation by principal investigator
- End of study

Survival follow-up

Every 3 months

Optional tumor biopsy prior to C2D1 in up to 80 patients

[a] The percentage of PD-L1–negative patients in cohort 1 will be limited to ~60%.
Primary prophylaxis with G-CSF will be prohibited in cycle 1, although therapeutic G-CSF will be allowed. Following completion of cycle 1, G-CSFs (prophylactic or therapeutic) will be permitted per standard guidelines. Erythropoiesis-stimulating agent administration and red blood cell transfusions will be allowed per investigator discretion.
C, cycle; D, day; GCb, gemcitabine plus carboplatin; G-CSF, granulocyte colony-stimulating factor; PD-1, programmed death protein-1; PD-L1, programmed death-ligand 1.

Optional Biopsy Collection

- To evaluate the impact of trilaciclib on changes to the tumor-associated immune response in TNBC, immunophenotypic changes will be compared between tumor biopsies from patients receiving trilaciclib or placebo prior to GCb
- For patients who consent to optional biopsy collection, fresh tumor biopsies from a recurrent/metastatic lesion will be collected at baseline and on-treatment, prior to cycle 2
 - Archival tissue is acceptable for use as the baseline sample if no systemic therapy or local radiation has been administered between biopsy and randomization
- Target participation for optional biopsy collection is 80 patients, including ~60 patients from cohort 1 and ~20 patients from cohort 2

Statistics

- Data from each cohort will be analyzed separately
- An interim analysis for OS will be performed for cohort 1 when ~70% of required events have been observed
- If the primary analysis of OS is statistically significant, then TTCD-fatigue will be analyzed

PATIENT ELIGIBILITY CRITERIA

Key inclusion criteria

- Adult patients (≥ 18 years of age)
- Confirmed locally advanced unresectable or metastatic TNBC
- No PD-1/PD-L1 inhibitor and no prior therapies in the metastatic setting (cohort 1); prior PD-1/PD-L1 inhibitor for ≥ 4 months' duration and documented PD-L1–positive status (cohort 2)
- Archival tumor tissue available, or fresh biopsy to be obtained
- ECOG performance status of 0 or 1
- Adequate organ function
- Resolution of nonhematologic toxicities from prior therapy to grade ≤ 1
- Predicted life expectancy of ≥ 3 months
- Vaccination against COVID-19 permitted

Key exclusion criteria

- Prior treatment with gemcitabine
- Prior treatment with carboplatin if completed ≤ 6 months prior to first metastatic recurrence
- Malignancies other than TNBC within 3 years prior to randomization
- Symptomatic CNS metastases and/or leptomeningeal disease requiring immediate treatment with radiation therapy or steroids
- Receipt of any cytotoxic chemotherapy or PD-1/PD-L1 inhibitor therapy (if relevant) ≤ 14 days prior to first dose of study drugs
- Known hypersensitivity to carboplatin or other platinum-containing compounds, or mannitol
- Pregnant or lactating women

CNS, central nervous system; ECOG, Eastern Cooperative Oncology Group; PD-1, programmed death protein-1; PD-L1, programmed death-ligand 1; TNBC, triple-negative breast cancer.

ENDPOINTS

Primary endpoint
Overall survival

Key secondary endpoint
TTCD-fatigue, as measured by FACIT-F

Other secondary endpoints
- OS in PD-L1–positive/negative subgroups (cohort 1 only)
- PFS
- Antitumor activity (ORR, CBR, TTR, DOR, and BOR, per RECIST v1.1)
- CIM-related symptoms and functional limitations
- Myeloprotection effects
- Safety and tolerability

Exploratory endpoints
- Pharmacodynamic effects in tumor and blood
- Antitumor efficacy by CDK4/6-dependence status

BOR, best overall response; CBR, clinical benefit rate; CDK4/6, cyclin-dependent kinase 4/6; CIM, chemotherapy-induced myelosuppression; DOR, duration of response; FACIT-F, Functional Assessment of Chronic Illness Therapy—Fatigue; ORR, objective response rate; OS, overall survival; PD-L1, programmed death-ligand 1; PFS, progression-free survival; RECIST, Response Evaluation Criteria in Solid Tumors; TTCD-fatigue, time to first confirmed deterioration of fatigue; TTR, time to response.

STUDY SITES

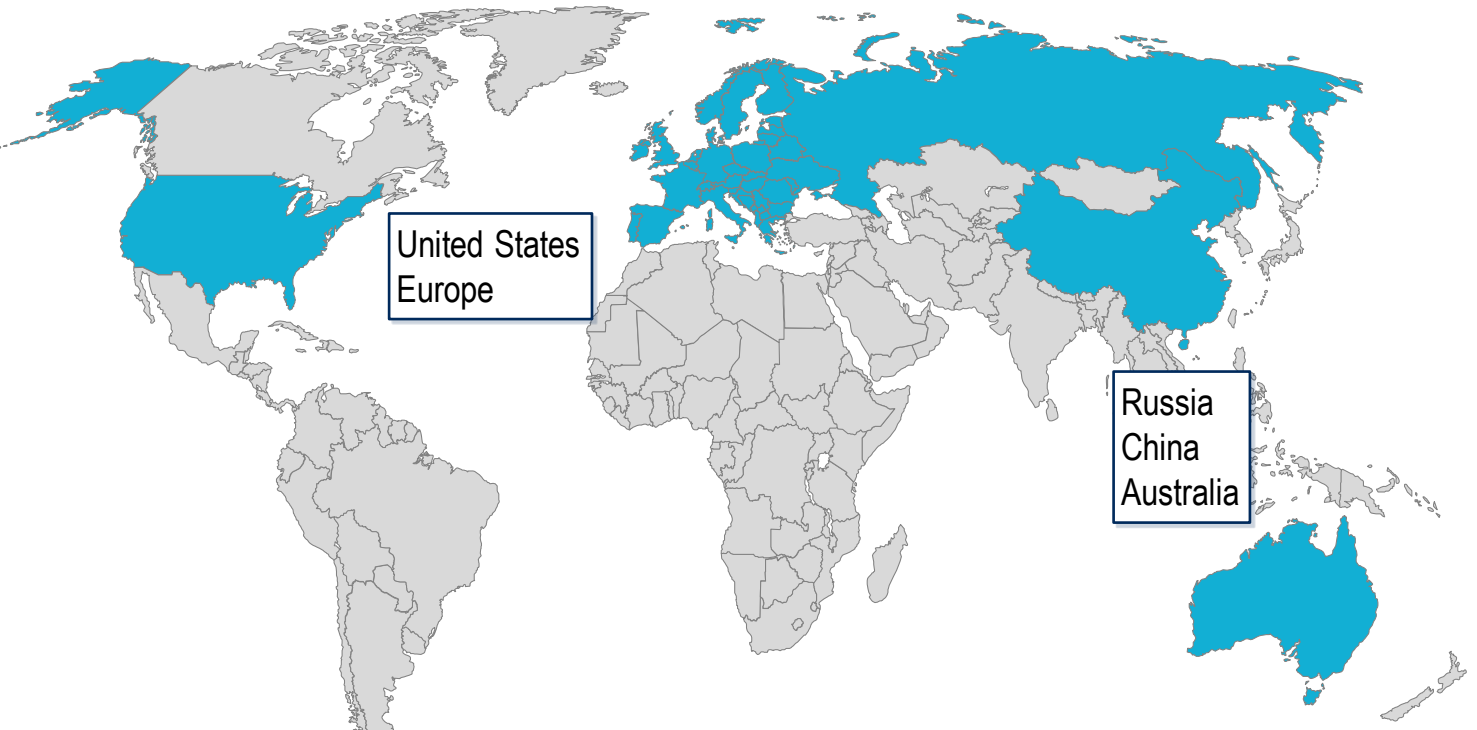

United States
Europe
Russia
China
Australia

Total study sites: 115
Estimated date first patient randomized: Q2 2021
Estimated date last patient completed: Q2 2024

American Society of Clinical Oncology Annual Meeting
June 4–8, 2021 | Chicago, IL, USA

REFERENCES:
1. Twelves C, et al. *Crit Rev Oncol Hematol*. 2016;100:74–87.
2. Cortes J, et al. *Lancet*. 2020;396:1817–28.
3. Schmid P, et al. *N Engl J Med*. 2018;379:2108–21.
4. Emens LA, et al. *JAMA Oncol*. 2019;5:74–82.
5. Zitvogel L, et al. *Nat Rev Immunol*. 2008;8:59–73.
6. COSELA™ (trilaciclib). Prescribing Information. https://www.g1therapeutics.com/cosela/pi/. Accessed April 2021.
7. Daniel D, et al. *Int J Cancer*. 2021;148:2557–70.
8. Weiss JM, et al. *Ann Oncol*. 2019;30:1613–21.
9. Hart LL, et al. *Adv Ther*. 2021;38:350–65.
10. He S, et al. *Sci Transl Med*. 2017;9:eaal3986.
11. Li C, et al. *Cancer Chemother Pharmacol*. 2021;87:689–700.
12. Lai A, et al. *J Immunother Cancer*. 2020;8:e000847.
13. Tan AR, et al. *Lancet Oncol*. 2019;20:1587–601.
14. O'Shaughnessy J, et al. SABCS poster presentation. 2020; abstract #PD1-06.

ACKNOWLEDGMENTS:
Study sponsored by G1 Therapeutics. Medical writing assistance was provided by Farhana Burnett, PhD, from Alligent Europe (Envision Pharma Group), funded by G1 Therapeutics.

DISCLAIMER:
This presentation is the intellectual property of the author/presenter. Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors. Contact them at shom.goel@petermac.org or sjain@g1therapeutics.com for permission to reprint and/or distribute.